FASN inhibition studies in preclinical tumor models identify biomarkers that align with in vitro and in vivo sensitivity to TVB-2640

Timothy S. Heuer, Richard Ventura, Kasia Mordec, Julie Lai, Joanna Waszczuk, Glenn Hammonds, Marie O' Farrell, Douglas Buckley and George Kemble

3-V Biosciences, Menlo Park, CA

3-V BIOSCIENCES™

Introduction

- **3-V Biosciences' first-in-class, oral FASN inhibitor is in Phase I clinical trials for the treatment of solid tumors**
- Fatty acid synthase (FASN) catalyzes the synthesis of palmitate from acetyl-CoA, malonyl-CoA, and NADPH
- Palmitate and palmitate-derived lipids function in vital cellular processes such as energy metabolism and cellular membrane biosynthesis
- Palmitate is conjugated directly to specific proteins as a mechanism to affect protein localization and activation
- FASN tumor expression has been found to be increased in a stage-dependent manner with high expression associated with diminished patient survival
- FASN activity promotes the tumorigenic capacity of cells by multiple mechanisms including enhanced macromolecular biosynthesis and glucose metabolism, cell growth and survival signal transduction, cellular stress response, and resistance to chemotherapeutics
- In vitro and in vivo studies in preclinical tumor models demonstrate that FASN inhibition reduces tumor cell proliferation and induces apoptosis
- Preclinical studies have discovered biomarker candidates and provided insight into the mechanisms of action that result in tumor growth inhibition and apoptosis

Results

In Vitro Sensitivity of Selected Tumor Cell Lines to FASN Inhibition

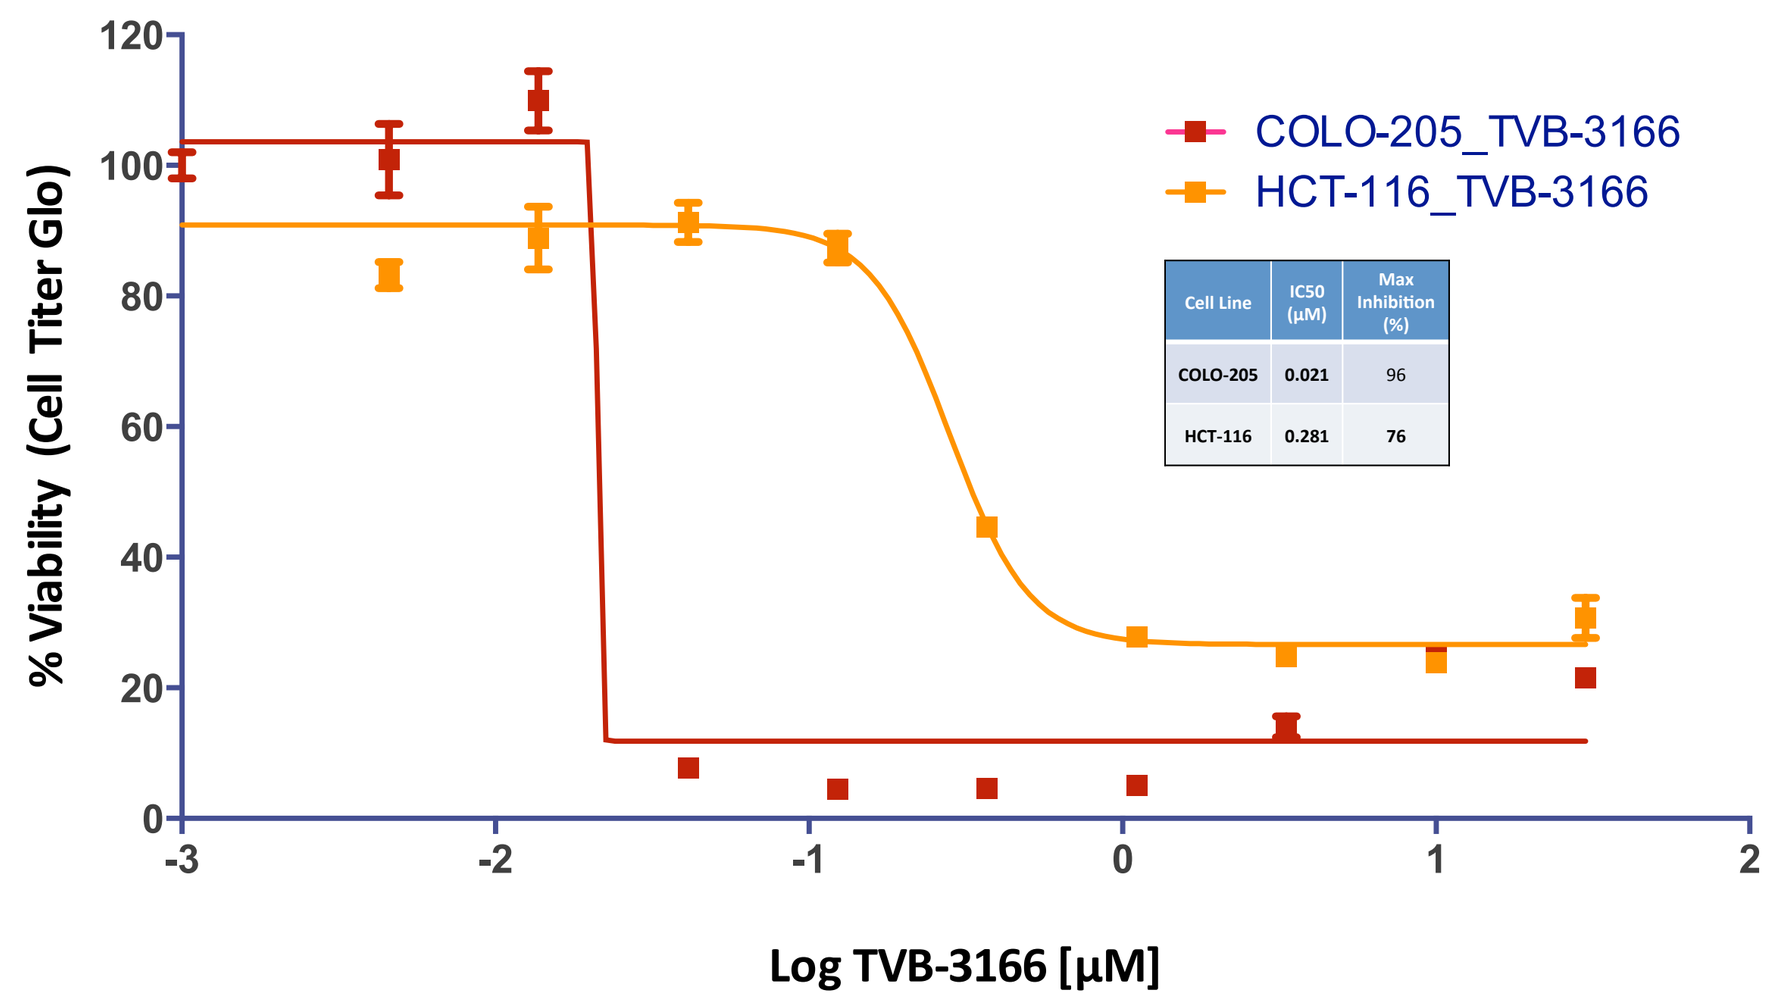
COLO-205_TVB-3166
HCT-116_TVB-3166
% Viability (Cell Titer Glo)
Log TVB-3166 [µM]
Cell Line | IC50 (µM) | Max Inhibition (%)
COLO-205 | 0.021 | 96
HCT-116 | 0.281 | 76

Baseline Beta-Catenin Expression Associates with FASN Inhibitor Sensitivity

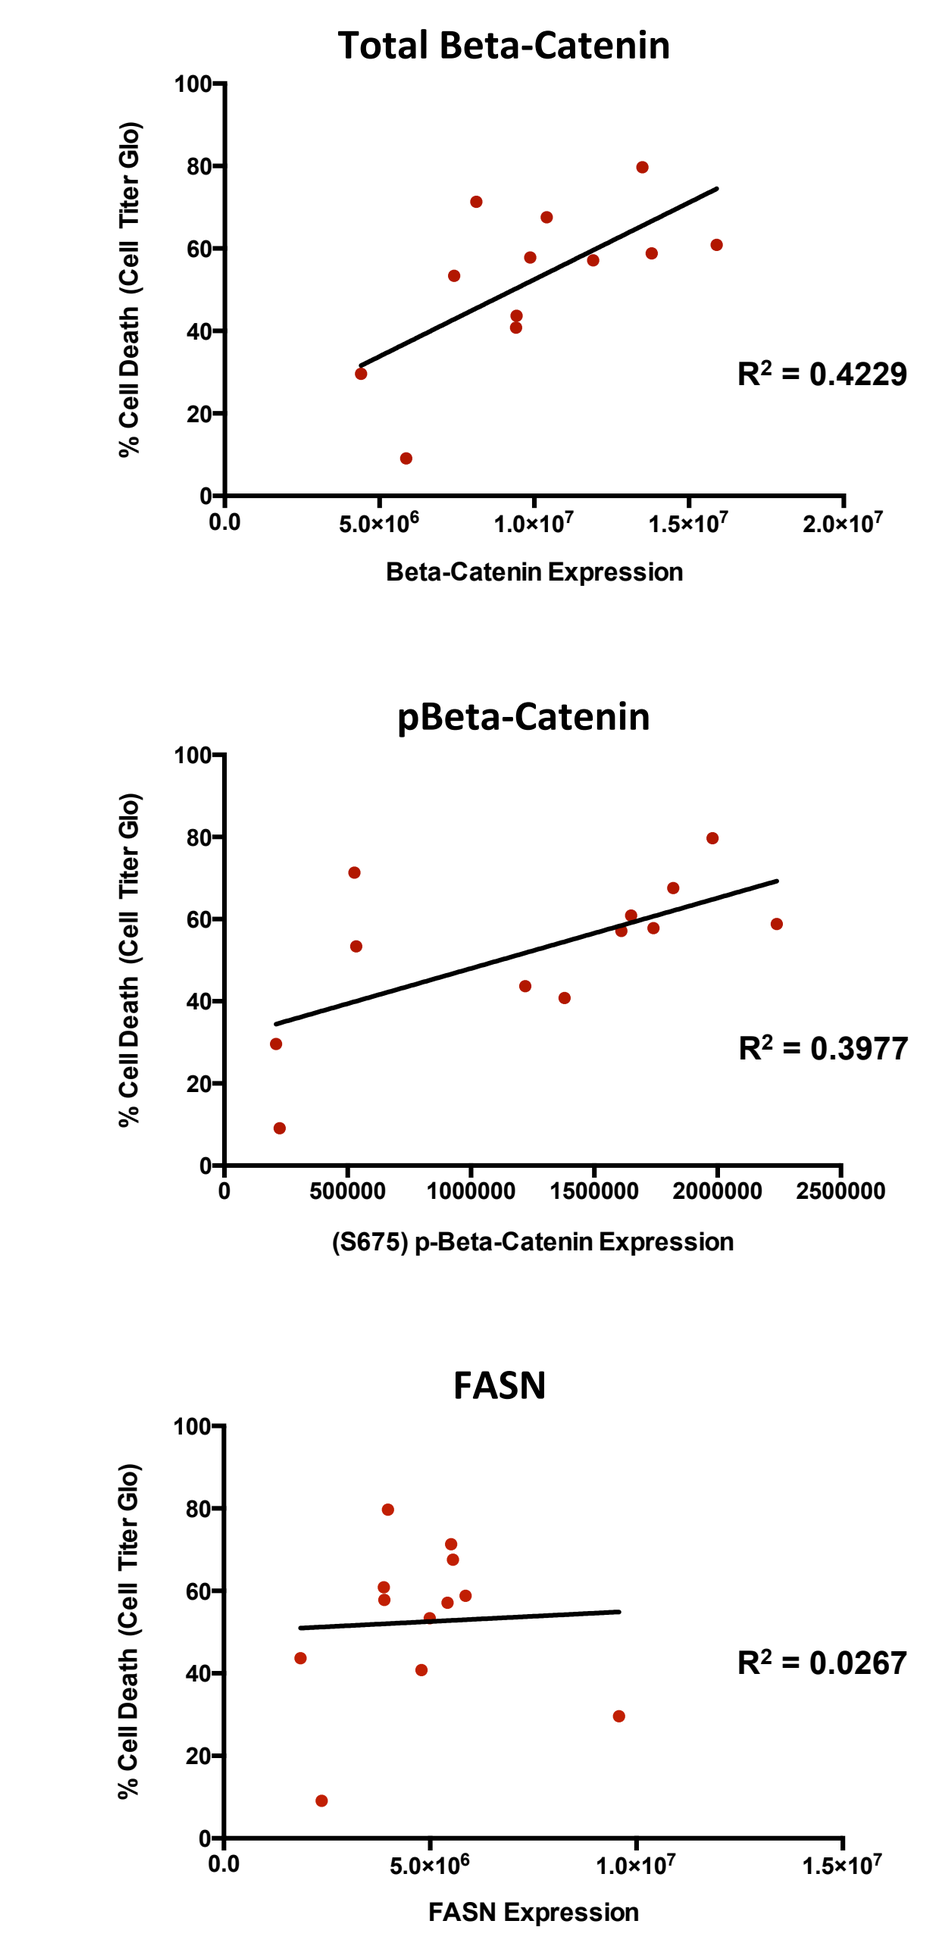
Total Beta-Catenin
% Cell Death (Cell Titer Glo)
Beta-Catenin Expression
R² = 0.4229
pBeta-Catenin
(S675) p-Beta-Catenin Expression
R² = 0.3977
FASN
FASN Expression
R² = 0.0267

Cell Line	7d CTG Inhibition (%)	FASN	LRP6	Beta-Catenin	pBeta-Catenin (S675)	cMyc	pAKT (S473)	α-tubulin
COLO-205*	86	ø	--	--	--	--	---	ø
T84*	77	ø	-	ø	ø	ø	--	ø
HCT-116	71	+	--	-	ø	--	+	ø
LS-1034	65	+	--	ø	-	--	---	-
OVCAR-8*	63	ø	-	ø	ø	ø	ø	ø
Lovo*	59	ø	-	ø	ø	-	-	ø
BT-549*	58	ø	--	-	--	--	-	ø
A549	54	+++	--	-	ø	-	-	ø
PC-3	37	-	--	-	-	-	ø	ø
LS123	23	+	-	ø	-	-	-	ø
SW-48	7	ø	-	-	-	-	ø	ø

2 µM or *0.2 µM TBV-3166

Legend		
	ø	No Effect
Increase	+	>20% - 100%
	++	>100% - 200%
	+++	>200%
Inhibition	-	>20% - 50%
	--	>50% - 75%
	---	>75%

Figure 1. In vitro characterization of tumor cell line sensitivity to FASN inhibition with TVB-3166. Cell Titer Glo assays were performed in Advanced MEM with 1%CS-FBS for 7 days. Western blot analysis was performed with cell lysates collected after 96 hours of treatment with TVB-3166 or 0.5% DMSO using the same media and serum conditions as for CTG assays.

TVB-2640 Inhibition of COLO-205 and HCT-116 Rat Xenograft Tumor Growth Aligns with In Vitro Sensitivity Data

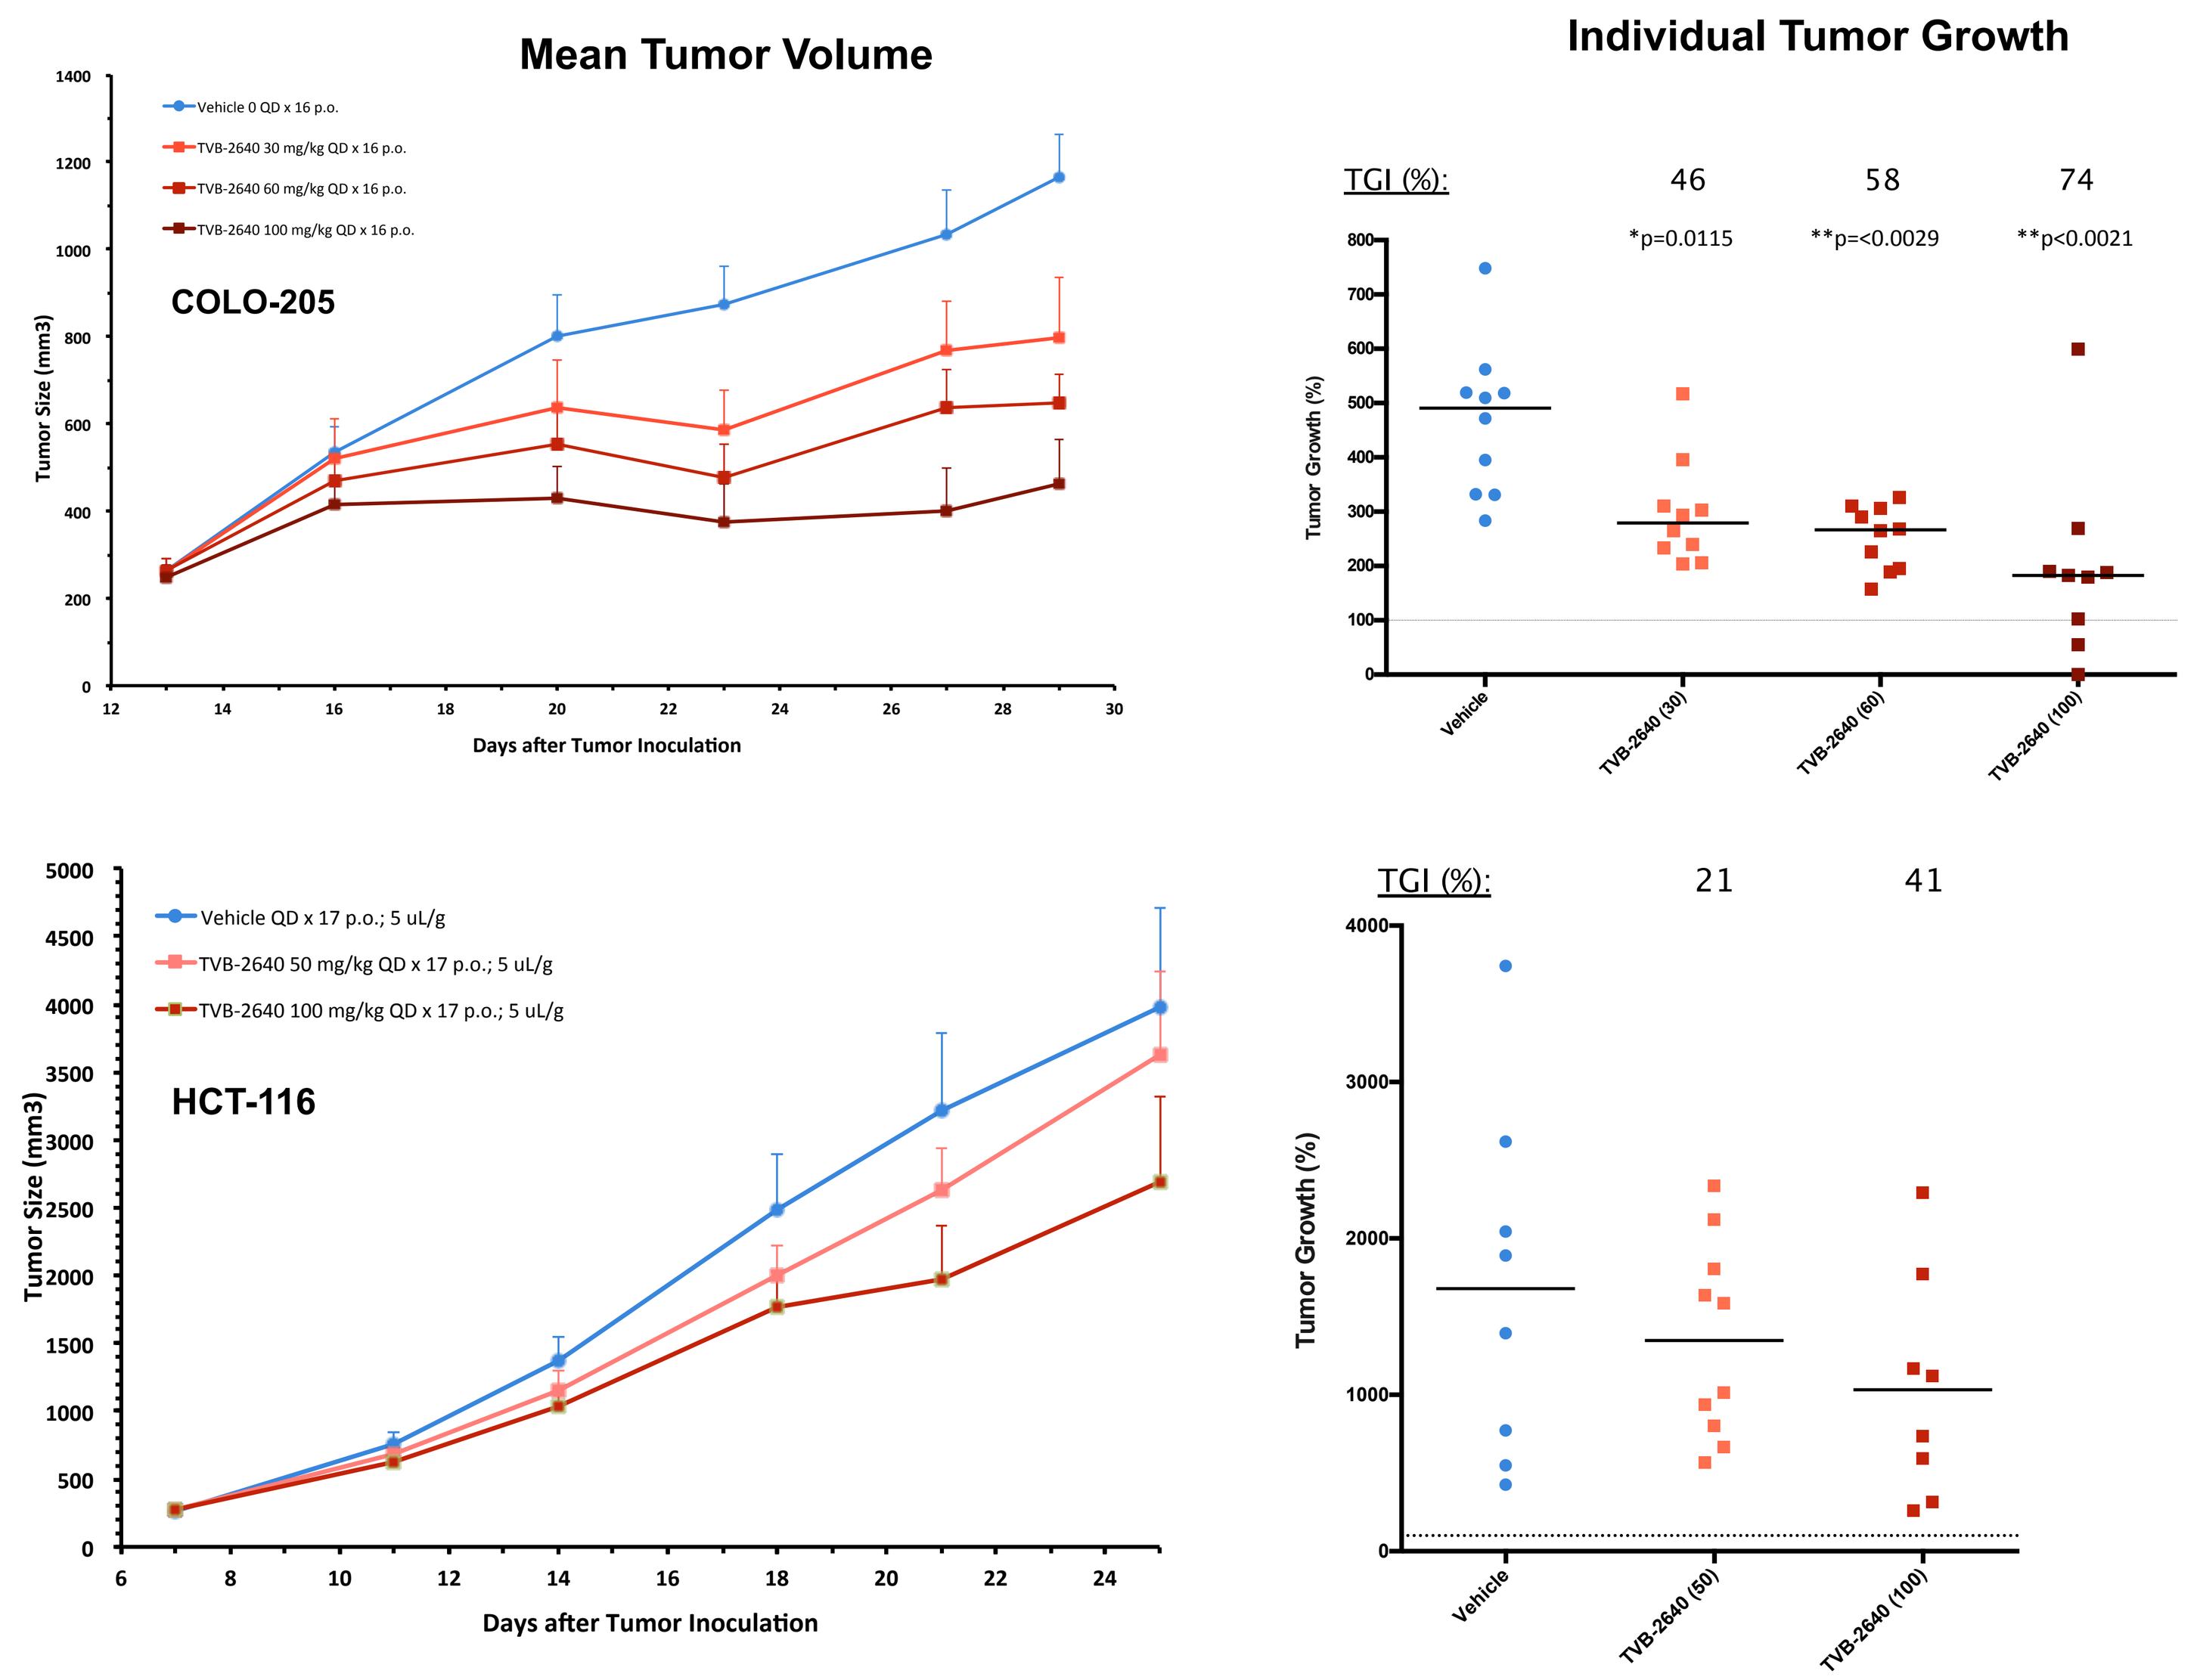
Mean Tumor Volume
Individual Tumor Growth
COLO-205
Vehicle 0 QD x 16 p.o.
TVB-2640 30 mg/kg QD x 16 p.o.
TVB-2640 60 mg/kg QD x 16 p.o.
TVB-2640 100 mg/kg QD x 16 p.o.
Tumor Size (mm3)
Days after Tumor Inoculation
TGI (%): 46 58 74
*p=0.0115 **p=<0.0029 **p<0.0021
Tumor Growth (%)
Vehicle
TVB-2640 (30)
TVB-2640 (60)
TVB-2640 (100)
HCT-116
Vehicle QD x 17 p.o.; 5 uL/g
TVB-2640 50 mg/kg QD x 17 p.o.; 5 uL/g
TVB-2640 100 mg/kg QD x 17 p.o.; 5 uL/g
TGI (%): 21 41
TVB-2640 (50)

Figure 2. Tumor growth analysis of of COLO-205 and HCT-116 tumors. Female Rowett nude rats (NIH-Foxn1^{rnu}), 10-12 weeks of age, were inoculated subcutaneously at the right flank with tumor cells (2 x 10^7) in 0.2 mL of PBS with matrigel (1:1). Tumor growth inhibition (TGI) was calculated as the percentage of tumor growth relative to tumor size at the start of treatment in drug-treated compared to vehicle-treated groups. The Mann-Whitney U test was used to assess statistical significance. Crown Biosciences (Santa Clara, CA; Beijing, China) conducted the in-life phase of the studies.

TVB-2640 Induces a Metabolic Signature of FASN Inhibition in COLO-205 Xenograft Tumors

- Elevated malonyl carnitine, decreased palmitic acid, and altered beta-oxidation among most significant changes observed by metabolomic profiling

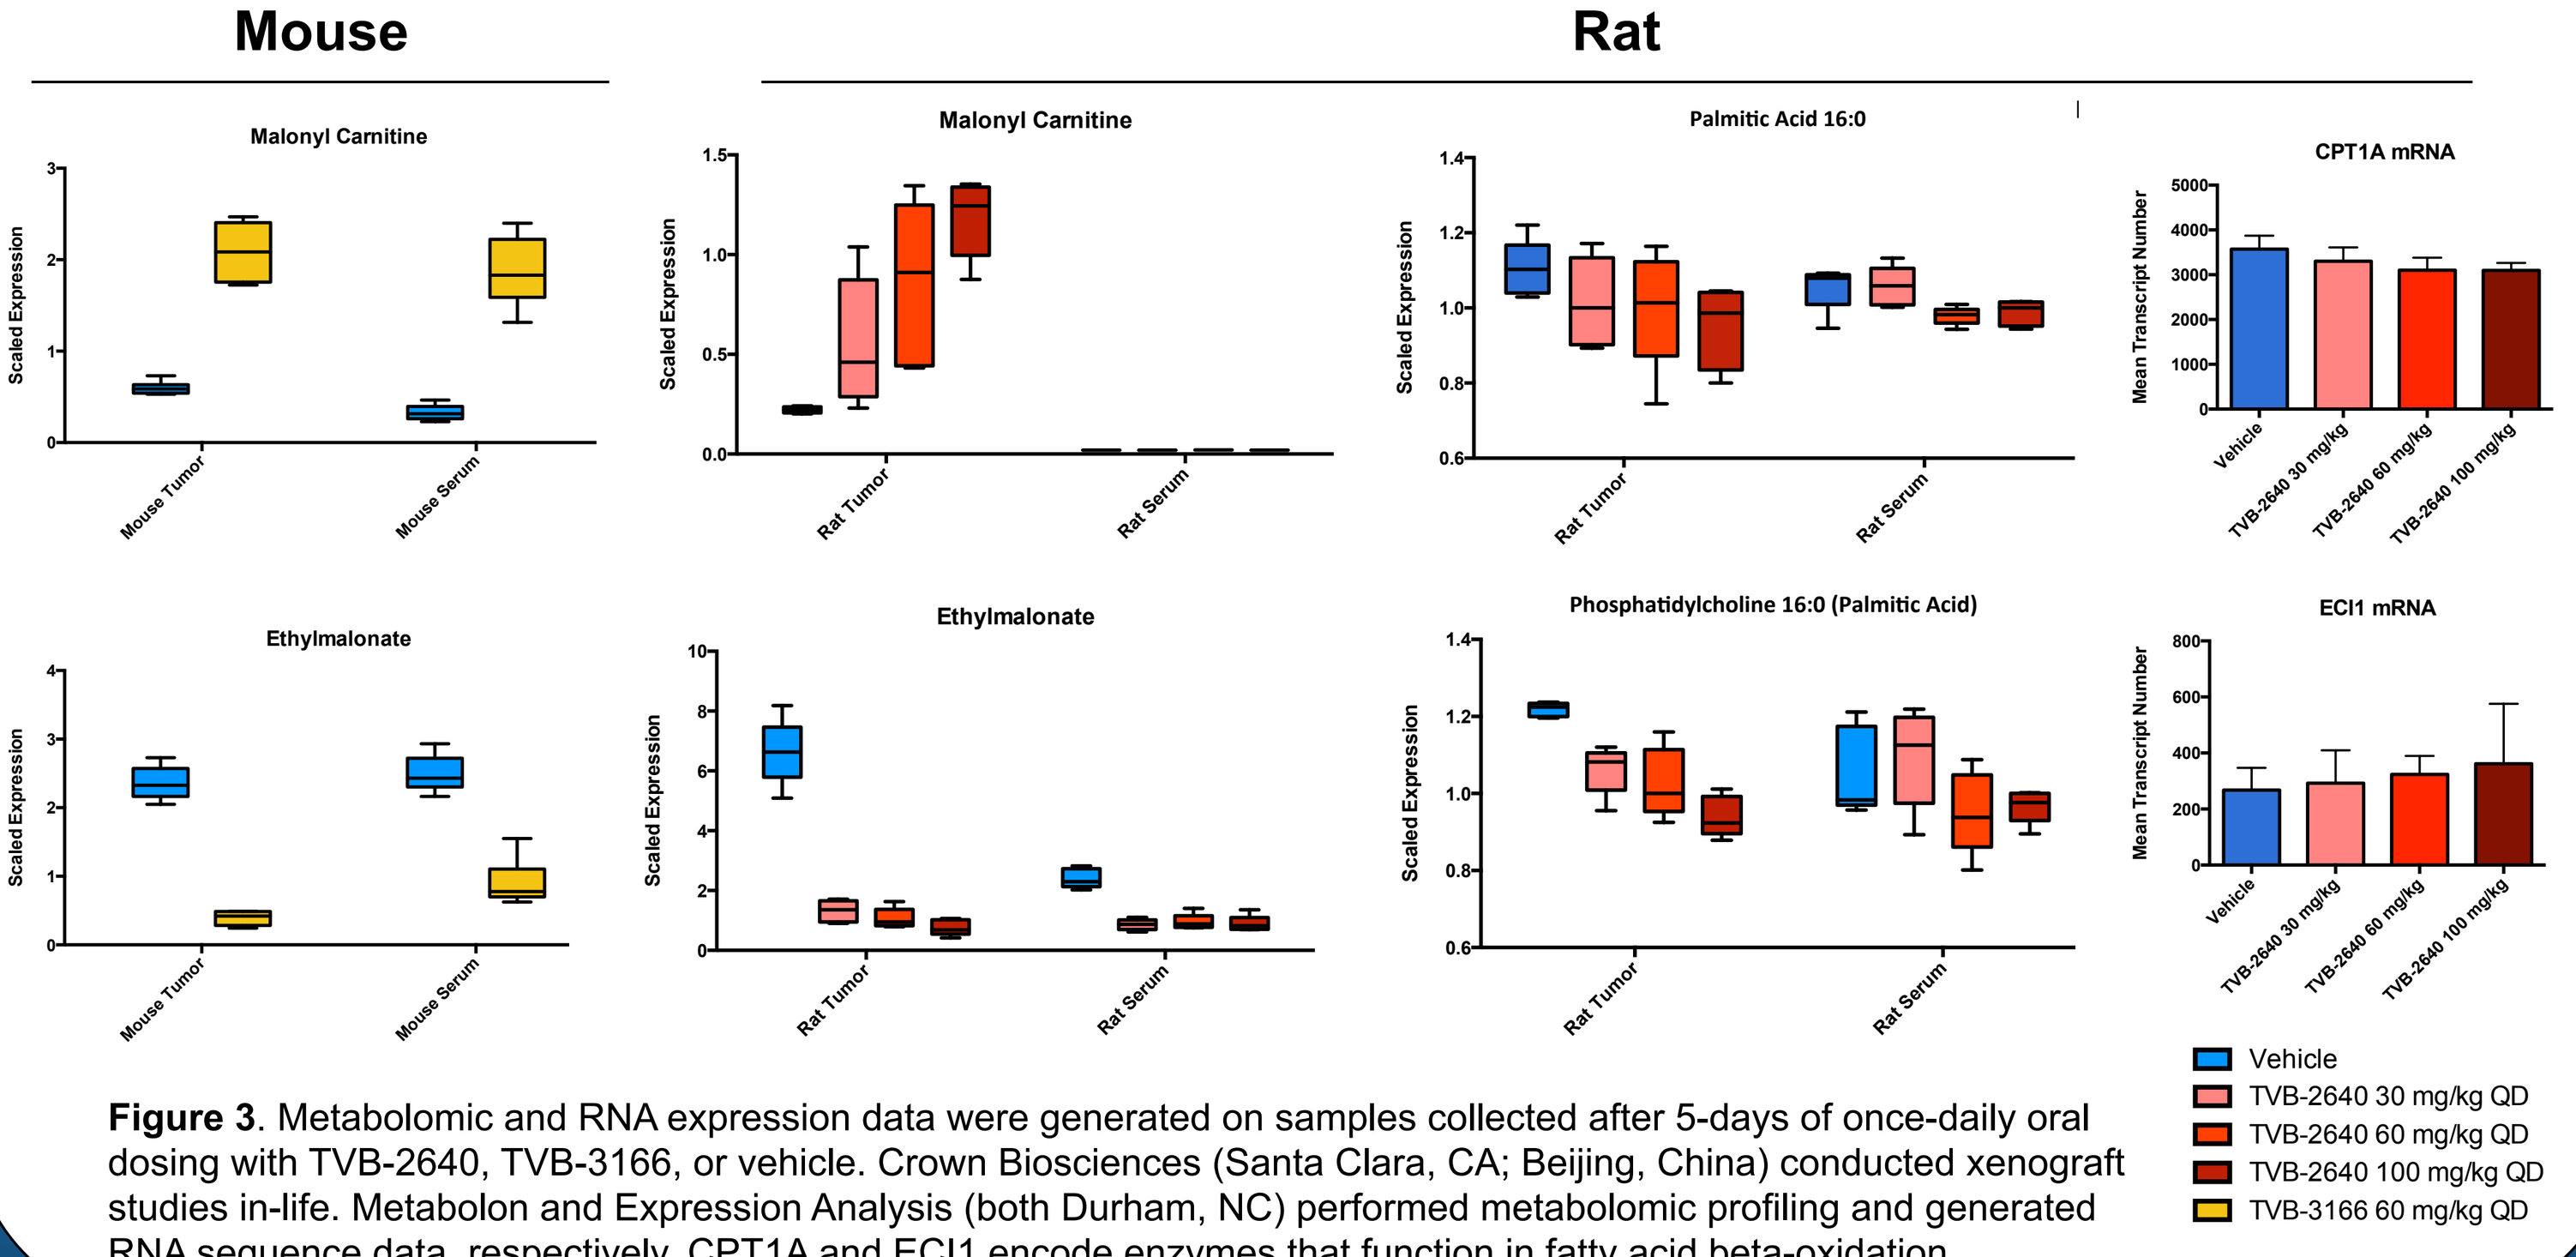
Mouse
Rat
Malonyl Carnitine
Ethylmalonate
Palmitic Acid 16:0
Phosphatidylcholine 16:0 (Palmitic Acid)
CPT1A mRNA
ECI1 mRNA
Scaled Expression
Mean Transcript Number
Mouse Tumor
Mouse Serum
Rat Tumor
Rat Serum
Vehicle
TVB-2640 30 mg/kg QD
TVB-2640 60 mg/kg QD
TVB-2640 100 mg/kg QD
TVB-3166 60 mg/kg QD

Figure 3. Metabolomic and RNA expression data were generated on samples collected after 5-days of once-daily oral dosing with TVB-2640, TVB-3166, or vehicle. Crown Biosciences (Santa Clara, CA; Beijing, China) conducted xenograft studies in-life. Metabolon and Expression Analysis (both Durham, NC) performed metabolomic profiling and generated RNA sequence data, respectively. CPT1A and ECI1 encode enzymes that function in fatty acid beta-oxidation.

TVB-2640 Induces Gene Expression Changes in Metabolic, Growth, Proliferation, and Survival Pathways in COLO-205 Xenograft Tumors, Less Significantly in HCT-116 Tumors

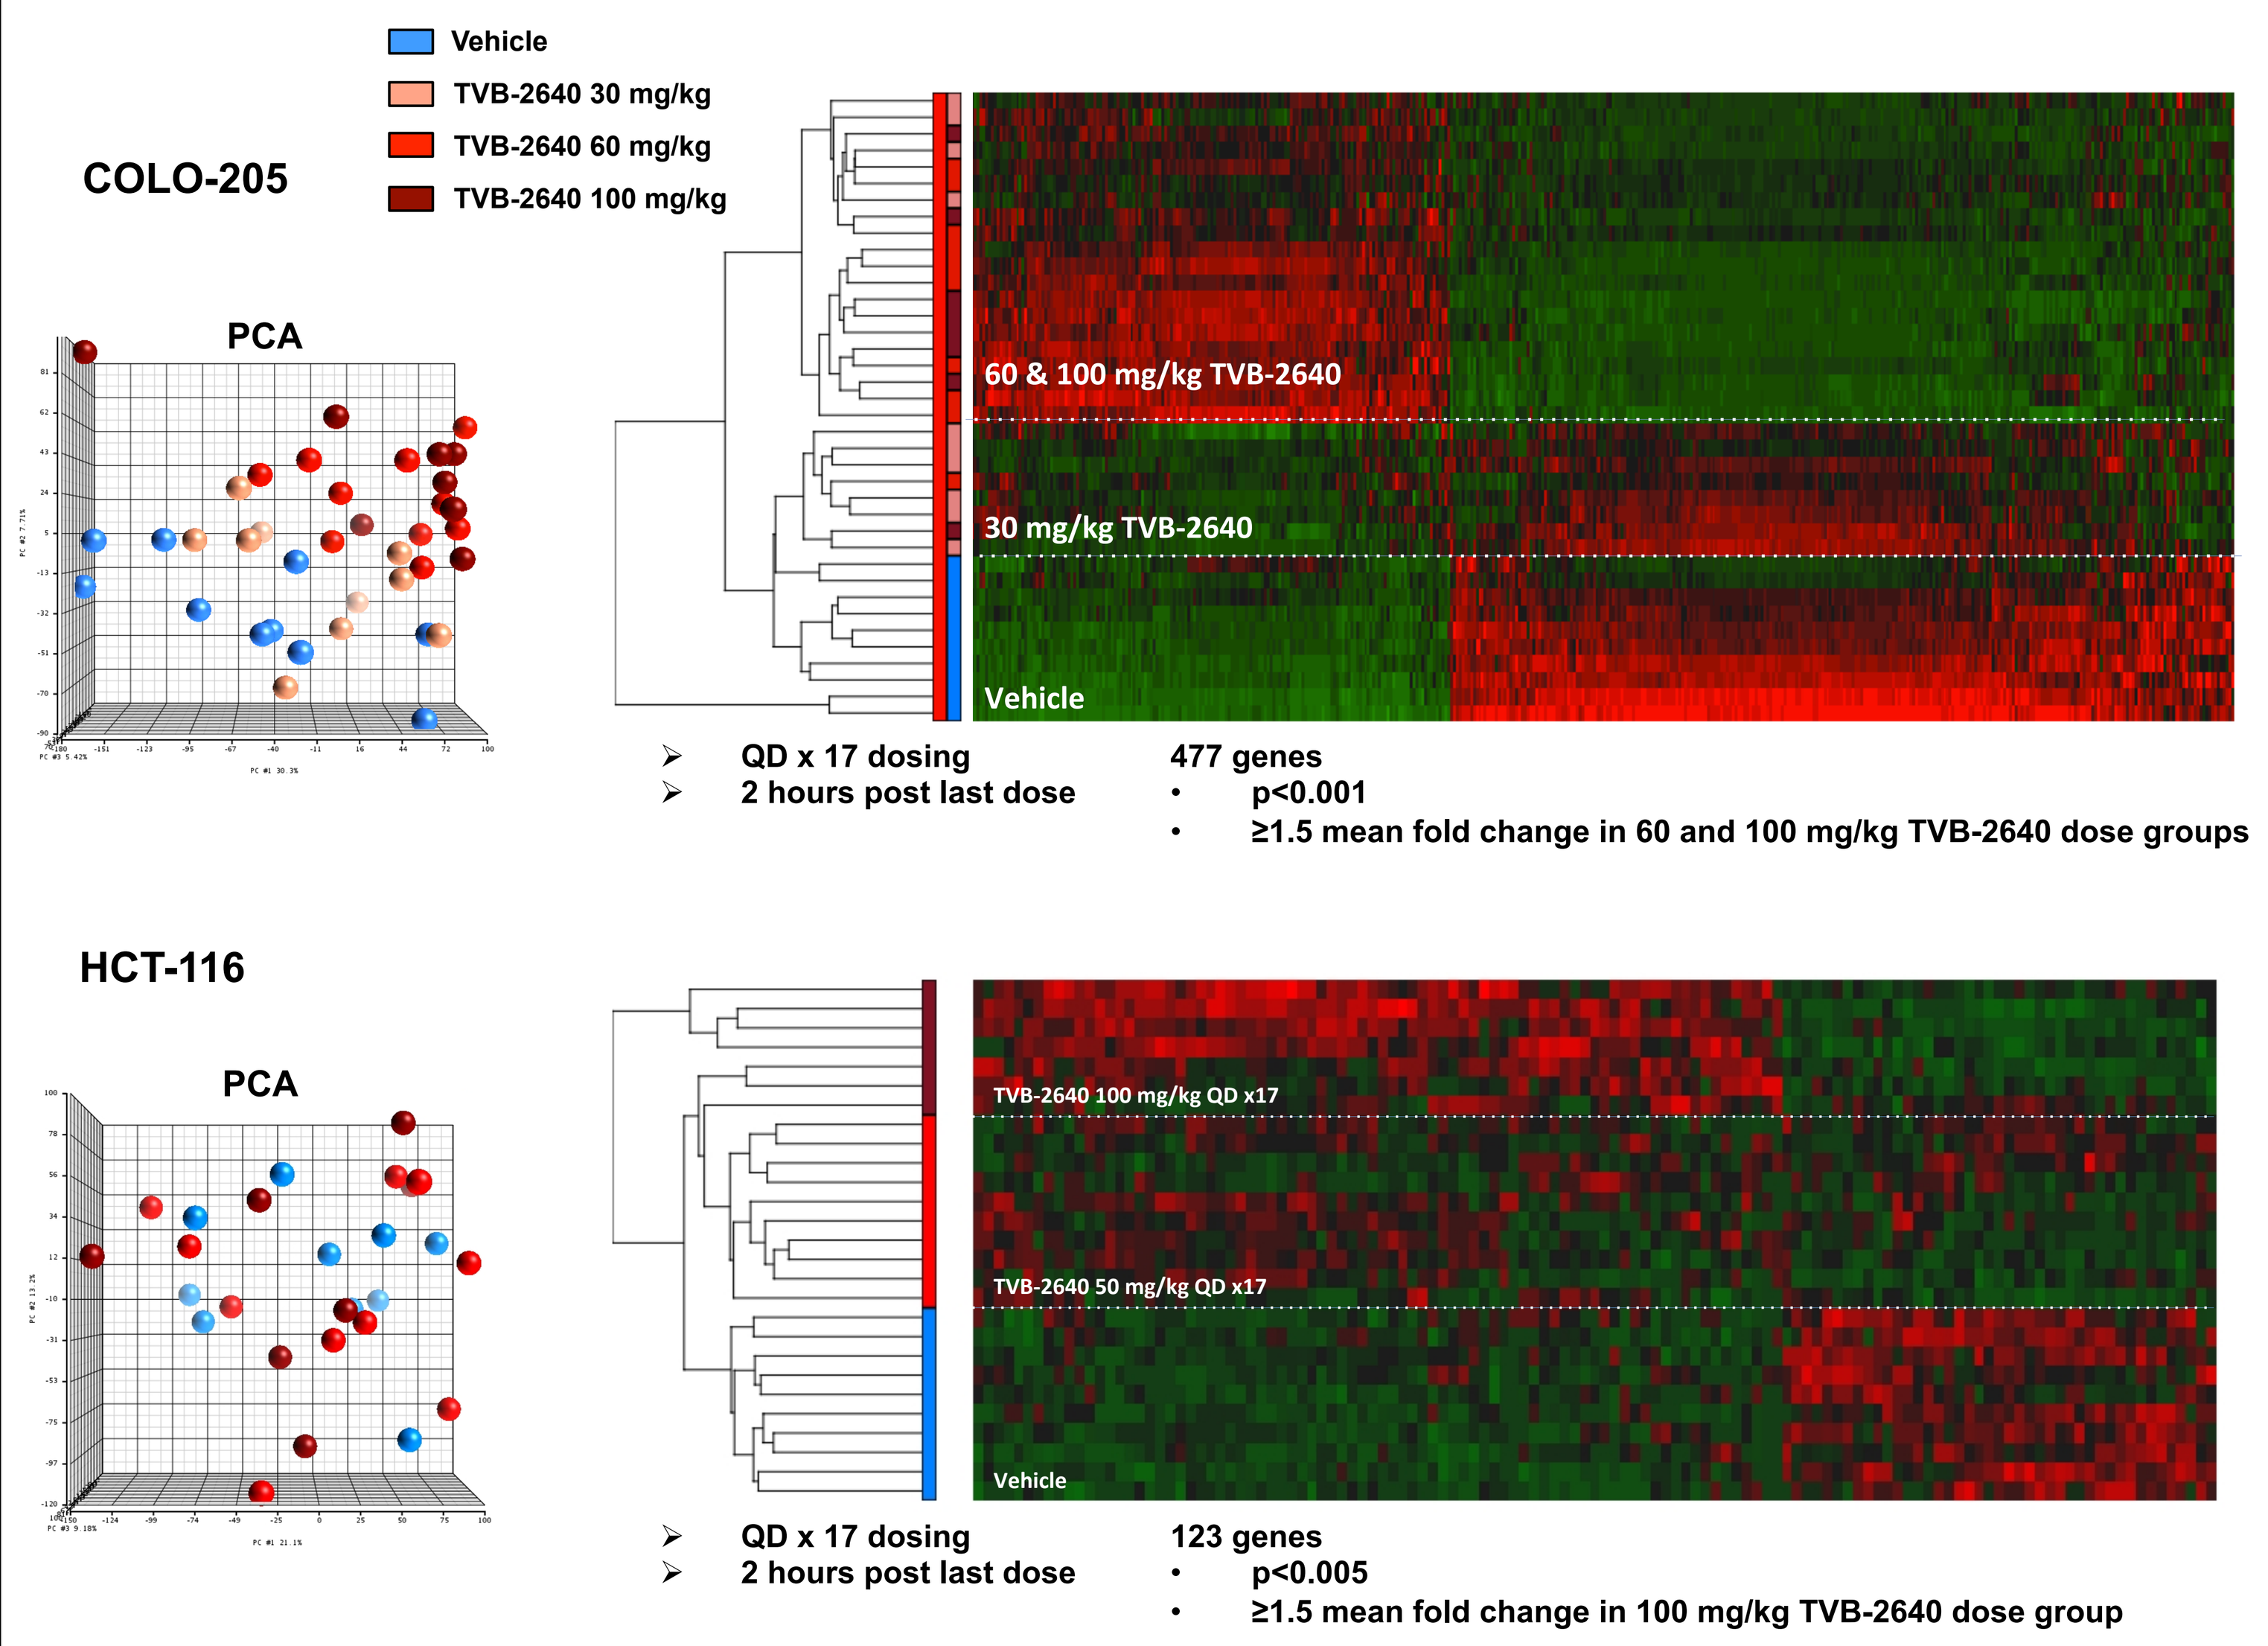
Vehicle
TVB-2640 30 mg/kg
TVB-2640 60 mg/kg
TVB-2640 100 mg/kg
COLO-205
PCA
60 & 100 mg/kg TVB-2640
30 mg/kg TVB-2640
Vehicle
QD x 17 dosing
2 hours post last dose
477 genes
p<0.001
≥1.5 mean fold change in 60 and 100 mg/kg TVB-2640 dose groups
HCT-116
PCA
TVB-2640 100 mg/kg QD x17
TVB-2640 50 mg/kg QD x17
Vehicle
QD x 17 dosing
2 hours post last dose
123 genes
p<0.005
≥1.5 mean fold change in 100 mg/kg TVB-2640 dose group

Figure 4. COLO-205 or HCT-116 rat xenograft tumors were harvested after 17 days of once-daily, oral treatment with TVB-2640 or vehicle (2 hours post final dose). RNA isolation and data analysis was performed at 3-V Biosciences. RNA sequencing (RNASeq-25, Illumina, Inc.) was performed by Expression Analysis (Durham, NC). Differential gene expression data analysis was performed at 3-V Biosciences using Partek Genomics Suite software (St. Louis, MO).

Inhibition of AKT and Beta-Catenin Pathways by TVB-2640 Associates with Efficacy in COLO-205 Xenograft Tumors

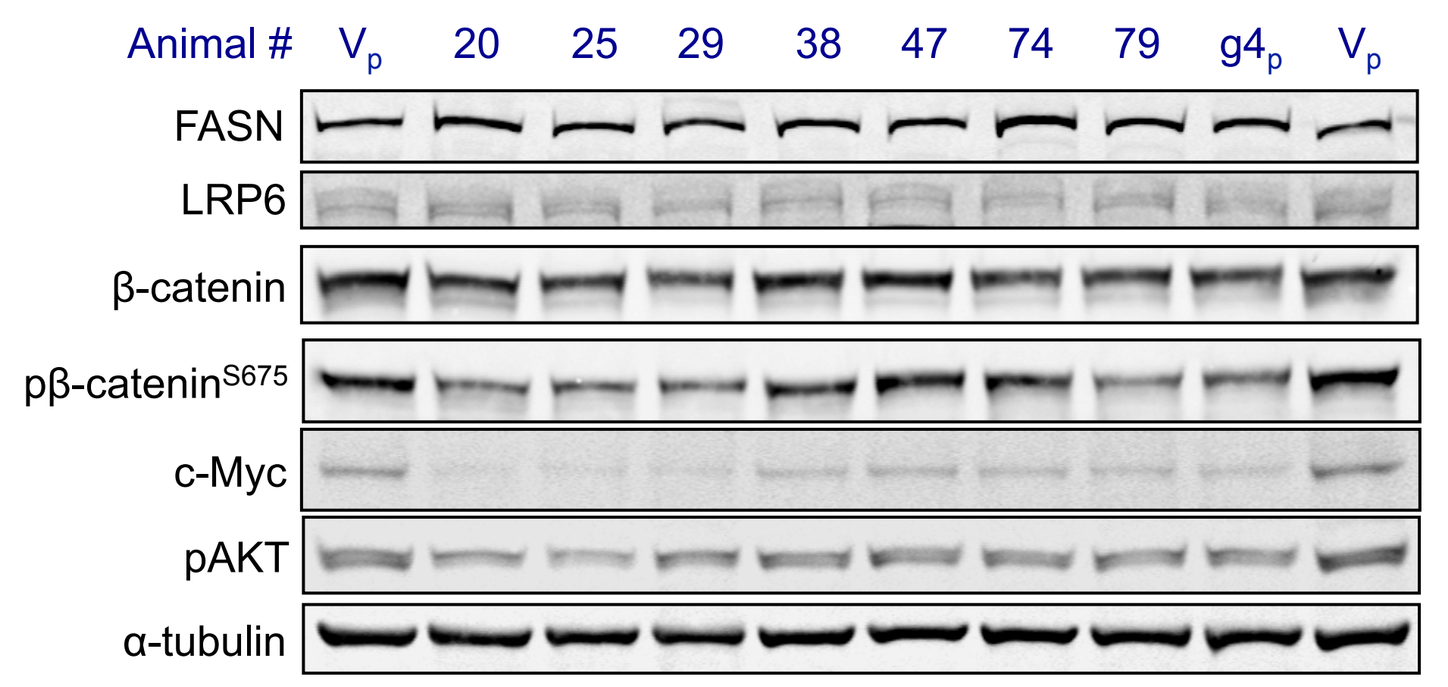
Animal # Vp 20 25 29 38 47 74 79 g4p Vp
FASN
LRP6
β-catenin
pβ-catenin S675
c-Myc
pAKT
α-tubulin

Animal #	V_p	20	25	29	38	47	74	79	$g4_p$	V_p
FASN	100	157	117	108	122	106	209	139	125	93
LRP6	100	85	68	54	78	56	61	67	57	95
B-catenin	100	82	68	62	86	81	72	76	78	99
pB-catenin	100	63	50	49	76	94	87	51	59	115
cMyc	100	21	20	19	48	56	38	32	37	126
pAKT (S473)	100	60	42	66	75	78	70	70	70	113
α-tubulin	100	96	85	80	100	96	105	97	97	105
% Tumor Growth	467	190	103	183	188	269	600	179		467
Tumor Size (mm³)		356	288	538	749	436	972	681		

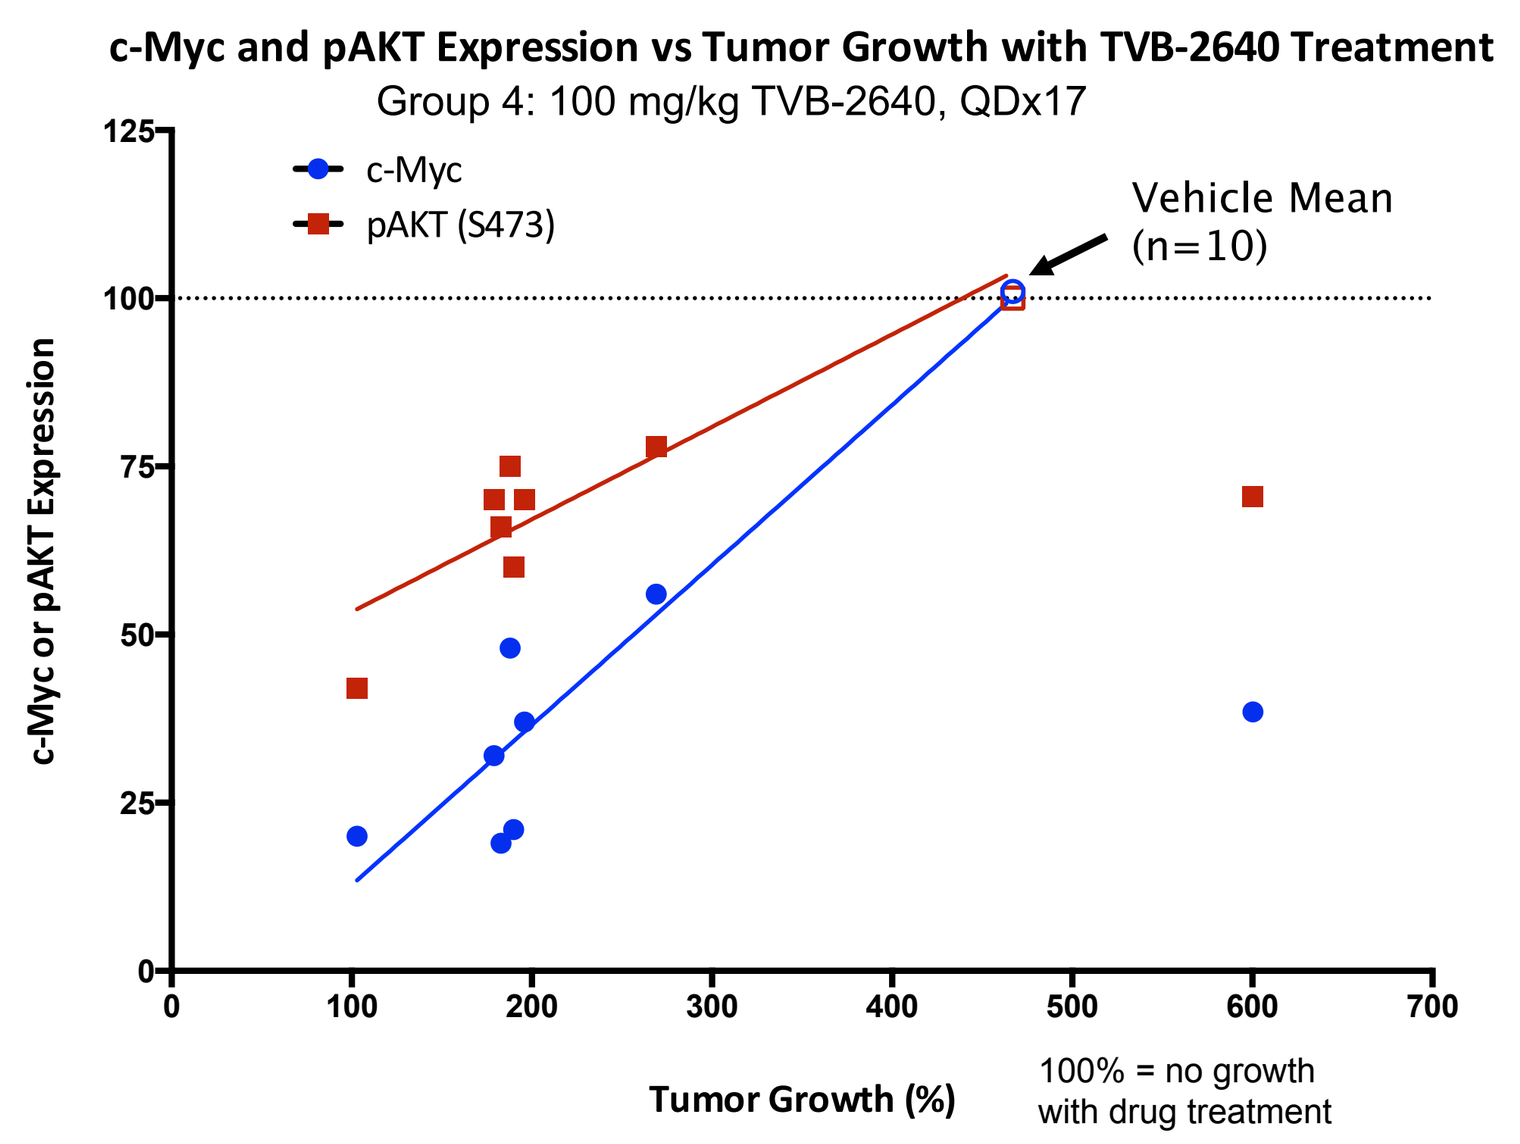
c-Myc and pAKT Expression vs Tumor Growth with TVB-2640 Treatment
Group 4: 100 mg/kg TVB-2640, QDx17
c-Myc
pAKT (S473)
Vehicle Mean (n=10)
c-Myc or pAKT Expression
Tumor Growth (%)
100% = no growth with drug treatment

Figure 5. COLO-205 rat xenograft tumors were harvested after 17 days of once-daily, oral treatment with TVB-2640 or vehicle (2 hours post final dose). Tumor lysate preparation and Western blot analysis were performed at 3-V Biosciences.

Gene Expression Signatures Classify Sensitivity of Tumor Cell Lines to FASN Inhibition

EMT[1] and lipogenic/glycolytic[2] signatures were applied to gene expression data in the CCLE for 102 tumor cell lines with 3-V FASN inhibitor sensitivity data

- FASN sensitivity associates with epithelial (classical), exocrine, and lipogenic classifications

Top quartile of FASN sensitive tumor cell lines by viability assay with TVB-3166

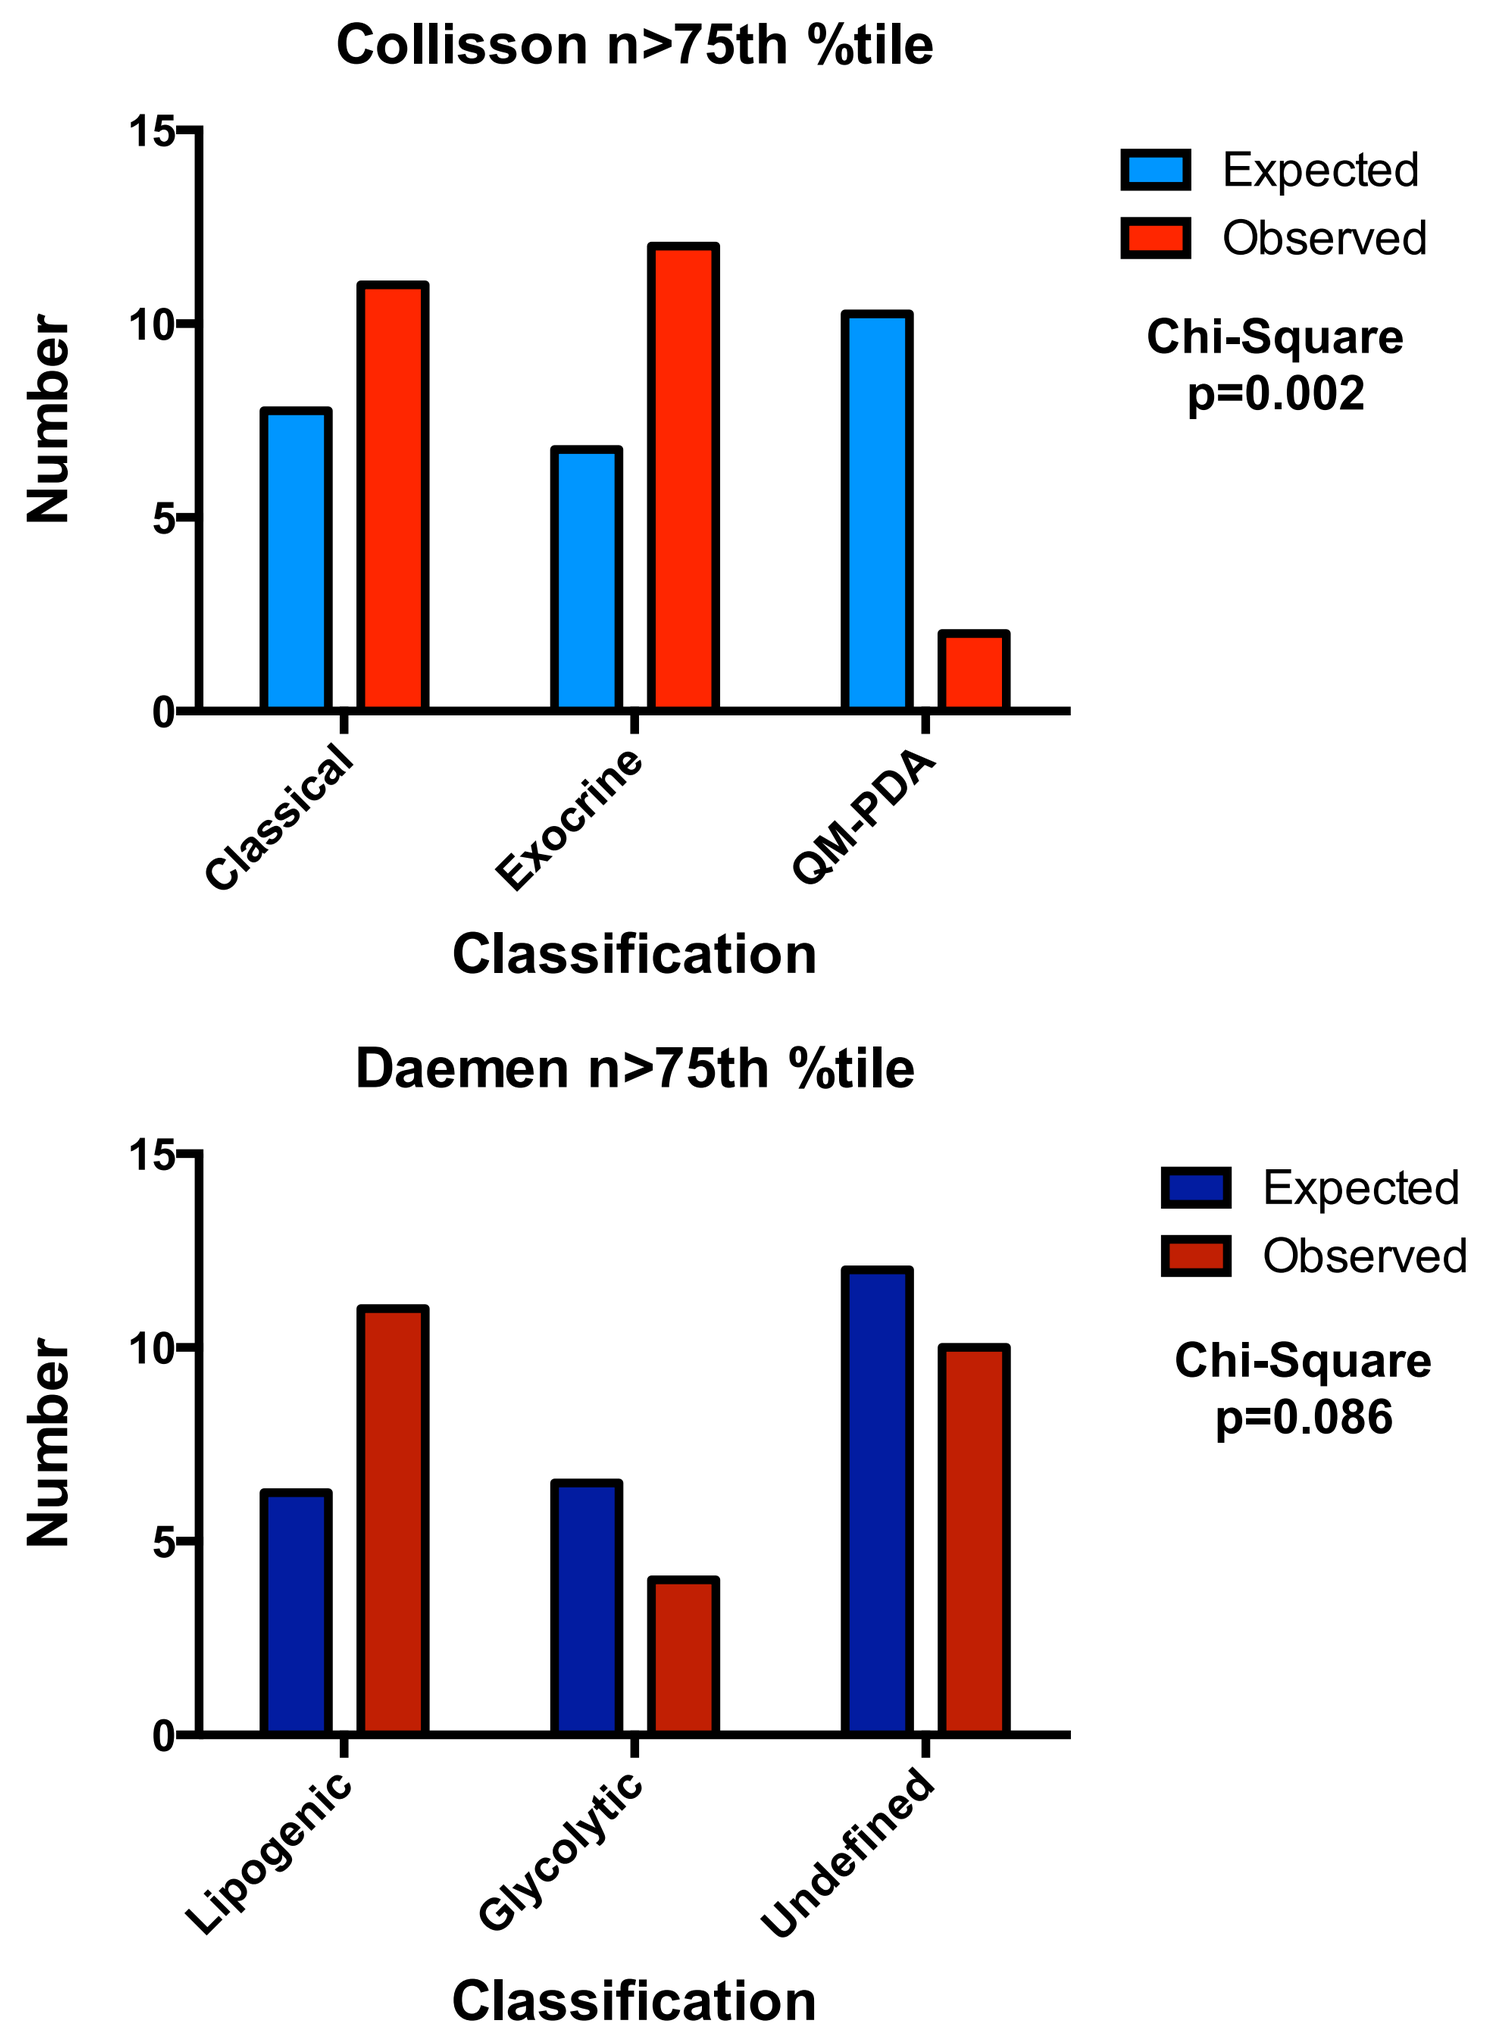
Collisson n>75th %tile
Expected
Observed
Chi-Square p=0.002
Number
Classical
Exocrine
QM-PDA
Classification
Daemen n>75th %tile
Expected
Observed
Chi-Square p=0.086
Lipogenic
Glycolytic
Undefined
Classification

Classification stratifies the cumulative distribution of tumor cell line sensitivity to FASN inhibition

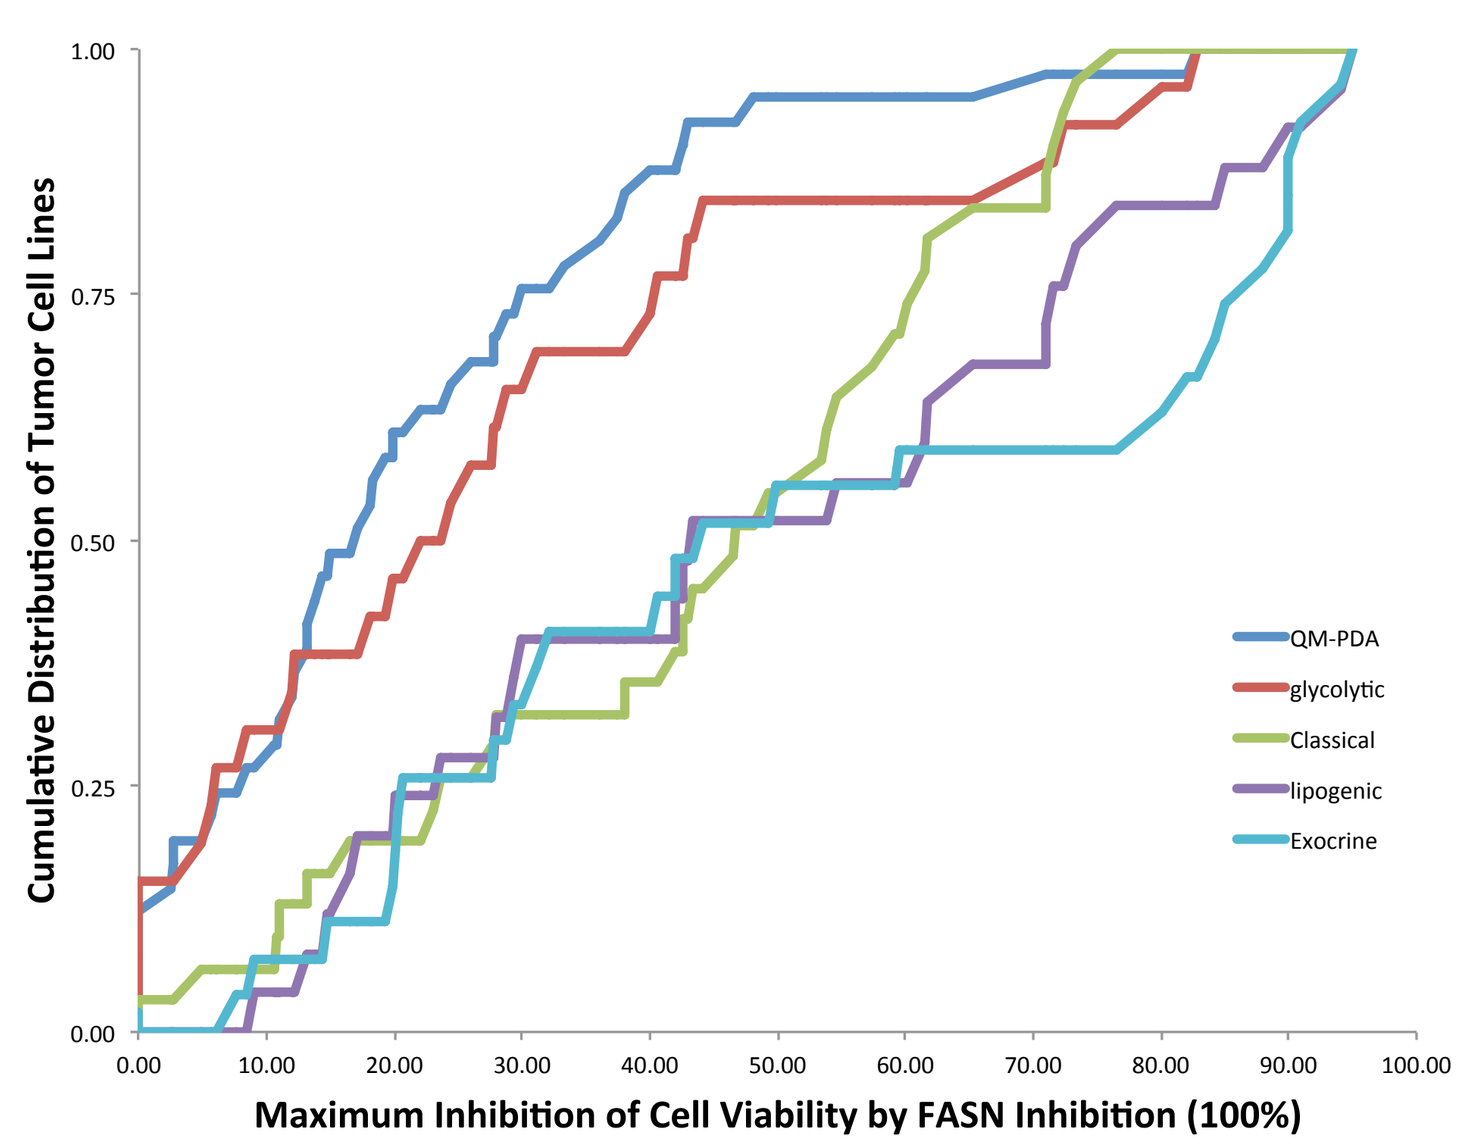
Cumulative Distribution of Tumor Cell Lines
Maximum Inhibition of Cell Viability by FASN Inhibition (100%)
QM-PDA
glycolytic
Classical
lipogenic
Exocrine

References

1. Collisson EA, Sadanandam A, Olson P, Gibb WJ, Truitt M, Gu S, Cooc J, Weinkle J, Kim GE, Jakkula L, Feiler HS, Ko AH, Olshen AB, Danenberg KL, Tempero MA, Spellman PT, Hanahan D, Gray JW.. Nat Med. 2011 Apr;17(4):500-3. doi: 10.1038/nm.2344. Epub 2011 Apr 3. PMID: 21460848

2. Daemen A, Peterson D, Sahu N, McCord R, Du X, Liu B, Kowanetz K, Hong R, Moffat J, Gao M, Boudreau A, Mroue R, Corson L, O'Brien T, Qing J, Sampath D, Merchant M, Yauch R, Manning G, Settleman J, Hatzivassiliou G, Evangelista M. Proc Natl Acad Sci U S A. 2015 Aug 11;112(32):E4410-7. doi: 10.1073/pnas.1501605112. Epub 2015 Jul 27. PMID: 26216984

Figure 6. 102 tumor cell lines representing diverse solid and hematopoietic tumor types were classified at 3-V using the gene sets and methods described in Collisson et al 2011 and Daemen et al 2015. FASN inhibitor sensitivity measured by the Cell Titer Glo assay using TVB-3166 was mapped onto the classifications. Chi-Square and cumulative distribution analyses showed enrichment of FASN-sensitive cell lines in the lipogenic, classical, and exocrine classes compared to glycolytic and QM-PDA classes. RNA expression data for the 102 cell lines was from the CCLE database (Broad Institute).

Conclusions and Status

- TVB-2640, a first-in-class oral FASN inhibitor, is in Phase I clinical development for the treatment of solid tumors.
- Expression of total beta catenin and S675 phosphorylated beta-catenin associate with sensitivity to FASN inhibition for many tumor cell lines.
- TVB-2640 demonstrates dose-dependent single agent tumor growth inhibition in the COLO-205 and HCT-116 rat xenograft tumor models with relative sensitivities that align with in vitro data.
- Tumor growth inhibition by TVB-2640 is associated with inhibition of β-catenin, c-Myc and AKT and modulation of tumor gene expression.
- Tumor cell line stratification according to published lipogenic, classical epithelial or exocrine classes enriches for in vitro FASN-inhibitor sensitivity compared to glycolytic or quasi-mesenchymal classification.
- Additional studies and analyses are ongoing to extend the discovery and assessment of biomarkers using tumor cell line and patient-derived tumor data.

printed by MegaPrint Inc. www.postersession.com